Simulating Defibrillation: Verification Using Defibrillation Thresholds and Surface Recordings

Jess D. Tate, BS, Jeroen G. Stinstra, PhD, Thomas A. Pilcher, MD, Ahrash E. Poursaid, Elizabeth V. Saarel, MD and Rob S. MacLeod, PhD.
University of Utah, Salt Lake City, UT

Introduction

Implantable cardioverter defibrillators (ICD) are considered a mature technology, but they are not optimized for use in pediatrics or patients with abnormal geometries [2], nor are they optimized to prevent excessive energy output, which can cause unnecessary damage [3].

We have developed a simulation to predict the electric field during the discharge of the ICD and calculate the energy required for defibrillation, or defibrillation threshold (DFT) [4-6]. DFTs observed clinically and predicted by the simulation; this approach also compares the surface potentials generated by the simulation with actual body surface potential measurements during ICD discharge.

Our simulations are patient specific and we compared clinically recorded DFTs and ICD potential maps to the corresponding simulated values for a cohort of four patients. Standard clinical testing provided the DFT values for each case. Potential maps were measured using a 32 lead system optimized for recording ICD potentials by applying a limited lead selection algorithm [7].

Methods

Limited Lead Selection: To capture the entire body surface potential maps from a finite set of leads that also avoided the sterile field and defibrillation patches, we carried out a statistical estimation appoach [7]. Training data from previous simulations provided the necessary information to derive a 32-lead system, which was tested using separate simulated test data (Figure 1).

Data Acquistion: Surface potentials using the 32-lead system and DFTs were acquired from four patients identified for ICD implantation and then compared to values predicted by the patient specific simulation (Figure 2). During the ICD testing, DFTs were found by sequentially increasing the shock energy until successful defibrillation, providing an minimum and maximum bounds for the actual DFT.

Patient Specific Models: were generated for each patient from a full torso, high resolution MRI or CT obtained prior to the ICD implantation. Segmentation of relevant organs and tissues (Seg3D) and numerical mesh generation (SCIRun) provided the necessary geometric models. The ICD was then electronically placed in the torso model as indicated by post-operation X-rays. The simulation pipeline was performed on each patient (SCIRun) to predict the surface potentials and DFTs. The predicted surface potentials were compared to the measured potentials using the metrics correlation (ρ), relative error (*RE*), and relative RMS error ($\bar{E}$). Seg3D and SCIRun can be downloaded from software.sci.utah.edu.

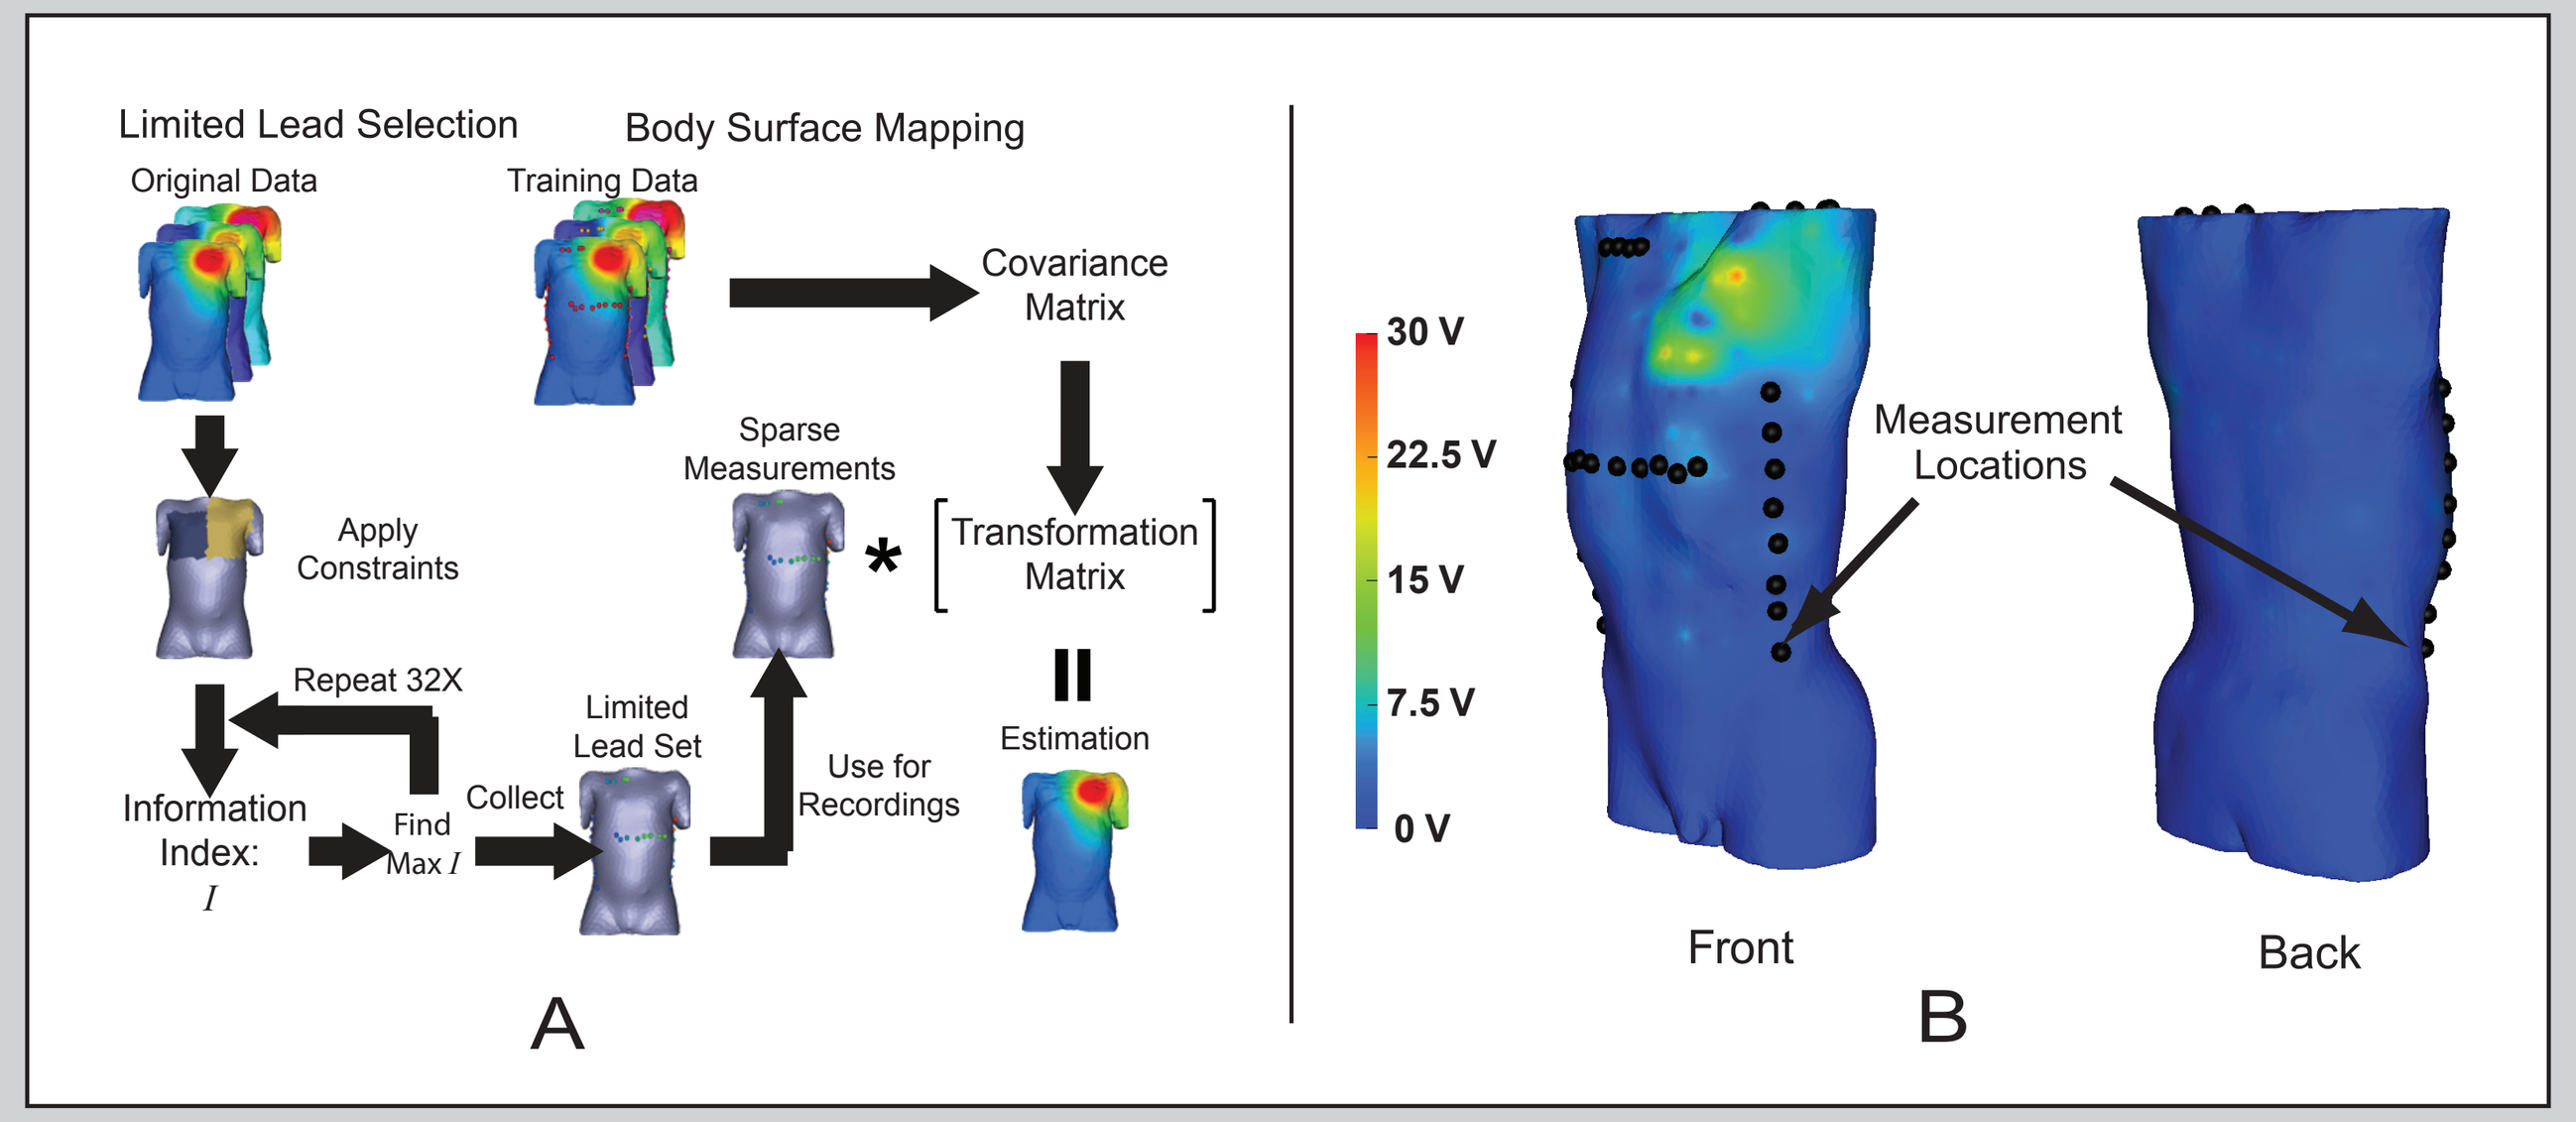
Limited Lead Selection
Body Surface Mapping
Original Data
Training Data
Covariance Matrix
Apply Constraints
Sparse Measurements
Transformation Matrix
Repeat 32X
Limited Lead Set
Use for Recordings
Estimation
Information Index: I
Find Max I
Collect
A
30 V
22.5 V
15 V
7.5 V
0 V
Measurement Locations
Front
Back
B

Figure 1. Limited lead selection algorithm implementation and typical error. A) Limited lead selection algorithm allows for full surface potential maps to be taken using only 32 lead locations [7]. B) Typical absolute error when simulated surface potentials are used as input into the optimized mapping system. Potentials are based on an ICD shock magnitude of 500 V.

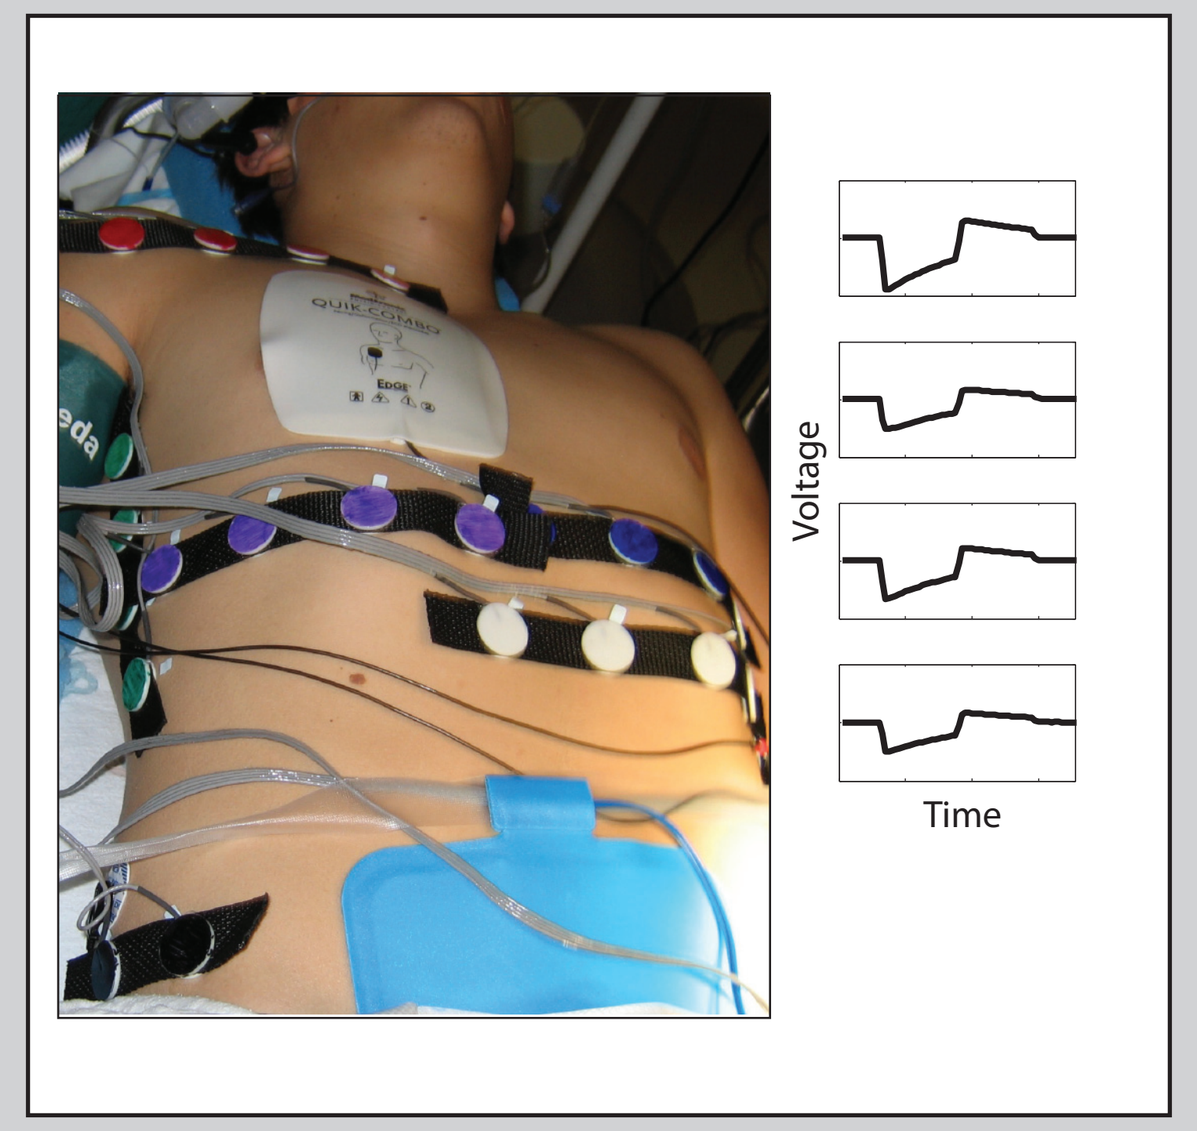
Voltage
Time

Figure 2. Obtaining body-surface potentials. 32 Surface leads were placed at pre-determined locations. Sample recordings are also shown.

Results

The mapping system developed for recording ICD surface potentials demonstrated high accuracy when tested on simulated potentials. The mean absolute error was 30 ± 20 V on a 500 V shock, correlation 0.9998 ± 3×10^{-4}, relative error 0.04 ± 0.06 %, and relative RMS error 0.6 ± 0.3 % (Figure 1). The level of accuracy was similar to previous applications of the algorithm [7].

Recorded and simulated discharge potentials were qualitatively and quantitatively similar indicating high overall accuracy. Figure 3 illustrates this, but there were noticeable differences apparent near the device location, i.e. the left shoulder. Summary statistics also show high accuracy with a mean correlation of 0.96 ± 0.02, relative error of 12 ± 5 %, relative RMS error of 7 ± 2 %. Individual shocks exhibited moderate to high accuracy (Figure 4b-d).

The simulation predicted two patient's DFTs within the clinical range and one just outside the clinical range. However, results from the 17 yo showed the largst differences between measured and simulated DFT values (Figure 4a). Interestingly, this patient also exhibited the highest error in the potential comparisons.

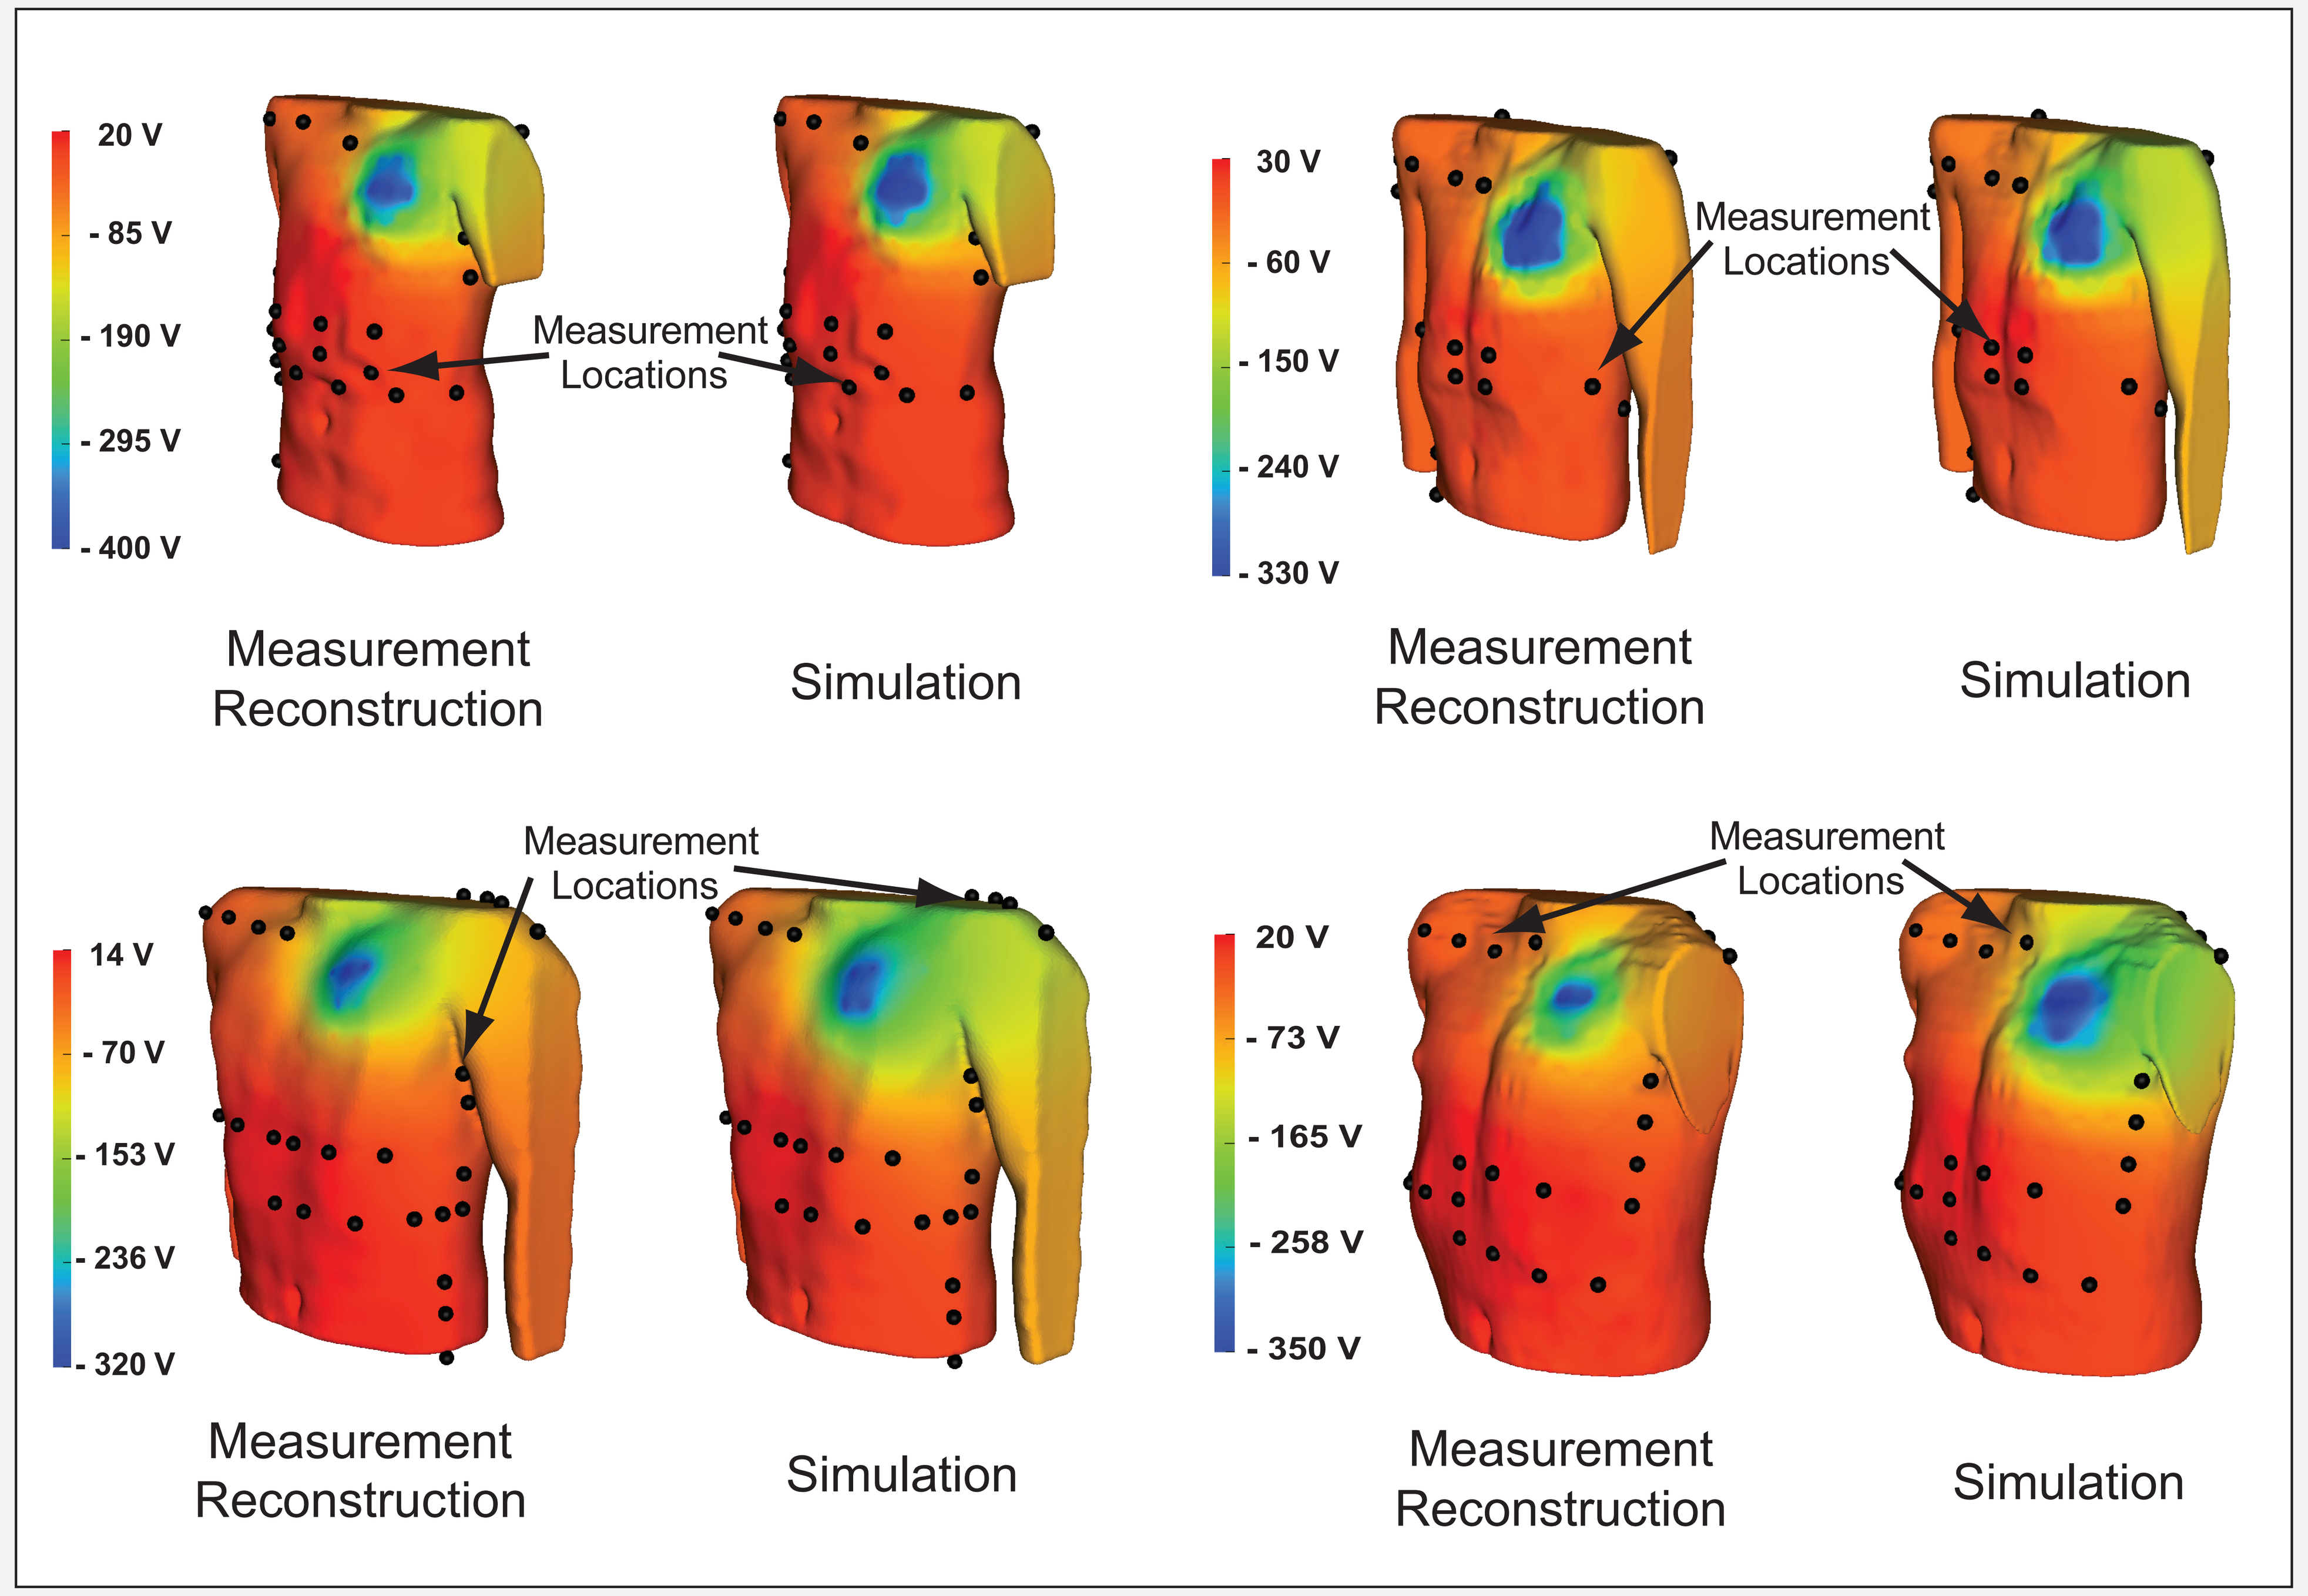
20 V
- 85 V
- 190 V
- 295 V
- 400 V
Measurement Locations
Measurement Reconstruction
Simulation
30 V
- 60 V
- 150 V
- 240 V
- 330 V
Measurement Locations
Measurement Reconstruction
Simulation
14 V
- 70 V
- 153 V
- 236 V
- 320 V
Measurement Locations
Measurement Reconstruction
Simulation
20 V
- 73 V
- 165 V
- 258 V
- 350 V
Measurement Locations
Measurement Reconstruction
Simulation

Figure 3. Sample comparisons of measured (left) and simulated (right) ICD discharge potentials. Each pair of potential maps corresponds to a separate subject.

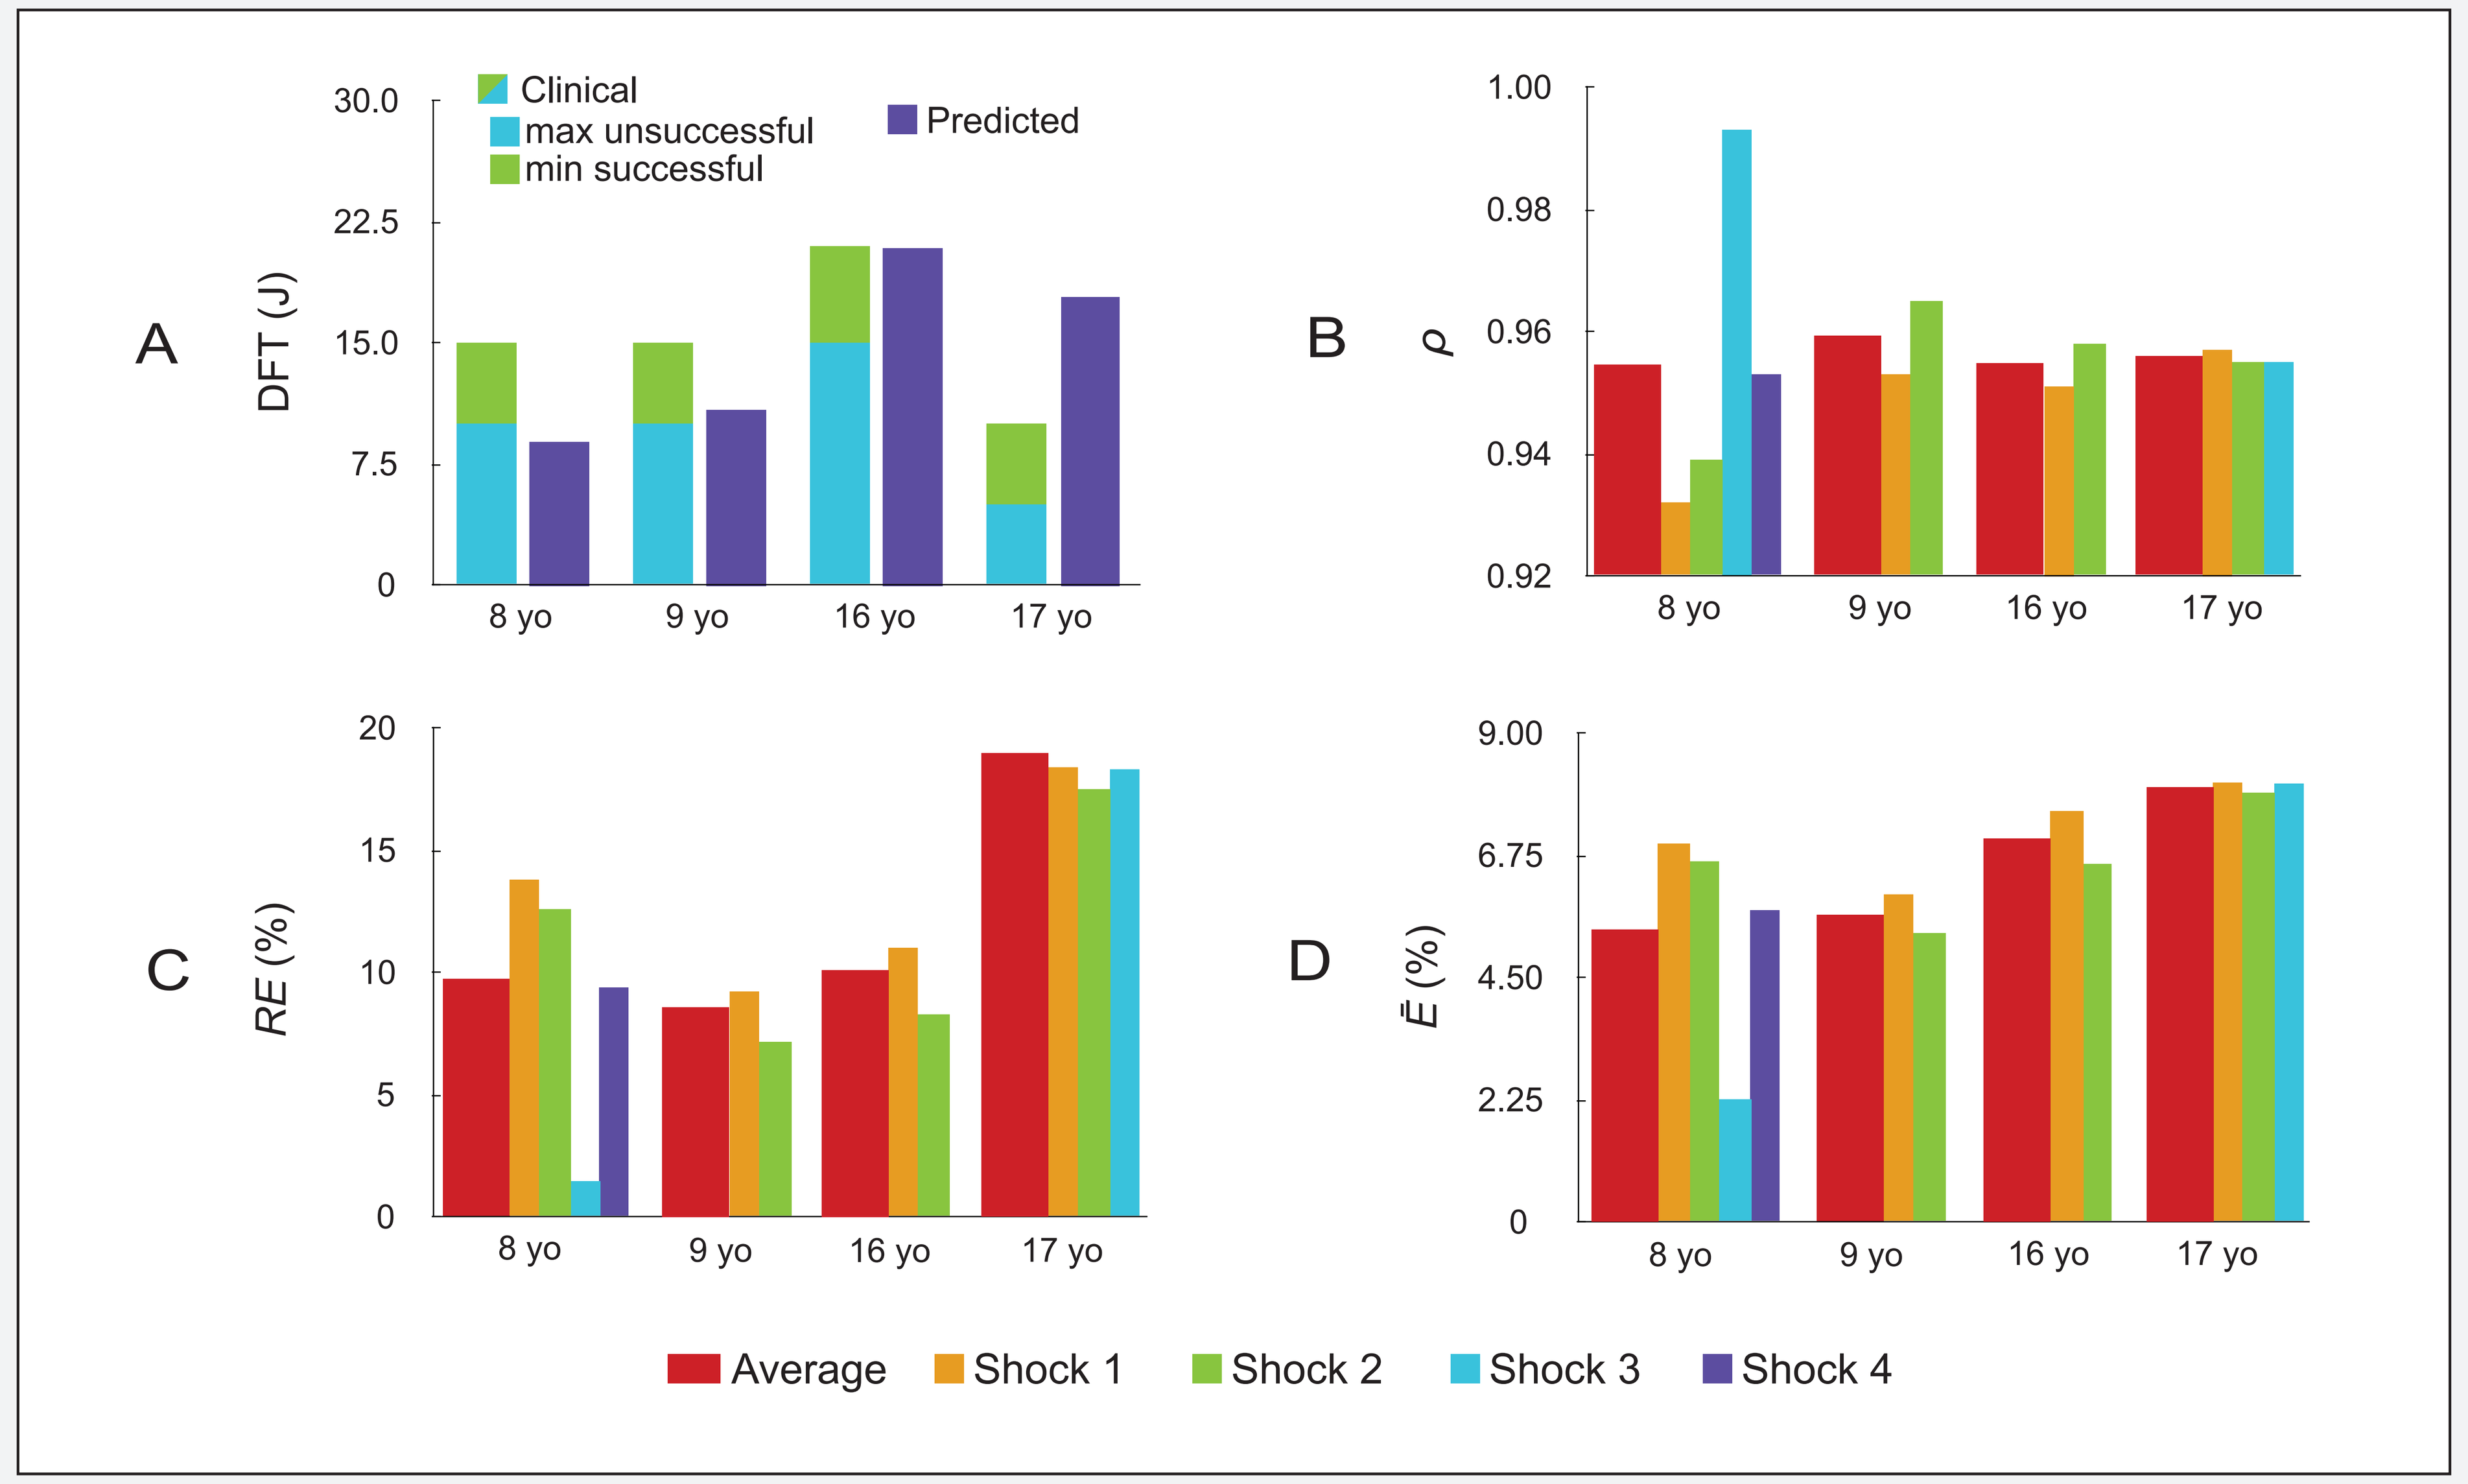
A
DFT (J)
Clinical
max unsuccessful
min successful
Predicted
30.0
22.5
15.0
7.5
0
8 yo
9 yo
16 yo
17 yo
B
ρ
1.00
0.98
0.96
0.94
0.92
C
RE (%)
20
15
10
5
0
D
Ē (%)
9.00
6.75
4.50
2.25
0
Average
Shock 1
Shock 2
Shock 3
Shock 4

Figure 4. Comparison of the DFTs and discharge potentials obtained clinically to predictions from the simulation. A) Clinically determined DFT ranges and the predicted DFT. B-D) metrics comparing simulated potential maps to those reconstructed from surface recordings. Comparisons between maps from individual shocks as well as the average for each patient are shown for the B) correlation (ρ), C) relative error (*RE*), and D) relative RMS error ($\bar{E}$).

Discussion

The 32-lead body surface mapping system, customized for this application, proved to be suitable means of acquiring discharge potentials during ICD testing.

Validation studies indicated a generally high level of agreement between measured and simulated discharge potentials (Figures 3&4), which adds confidence to the simulation and provides support for previous findings obtained using the pipeline [4-6].

Though this validation study showed high overall accuracy, it also suggests areas for improvement. The patient with greatest DFT error correlated with the highest surface potential error (Figure 4), indicating that an increase in accuracy of the potential field distribution may increase accuracy of the predicted DFT. One possible source of error is the inaccurate representation of the conductivities of tissues. A previous study [8], as well as our preliminary explorations in conductivities suggests that small changes in the conductivity values, especially of the myocardium and blood, can significantly alter the potential distribution. Another source of error is the assumption of isotropic, passive conductivity of the heart, which ongoing studies seek to resolve [9].

Financial Disclosure

Jess Tate—Nothing to disclose
Jeroen Stinstra—Nothing to disclose
Thomas Pilcher—Nothing to disclose
Ahrash Poursaid—Nothing to disclose
Elizabeth Saarel—Nothing to disclose
Rob MacLeod—Nothing to disclose

Acknowledgements

This research is made possible with support from the Primary Children's Medical Center Foundation Grant, the NIH/NCRR Center for Integrative Biomedical Computing, 2P41 RR0112553-12, and the Cardiovascular Research and Training Institute. Support was also provided by Matthew Jolley from Stanford University, Dana Brooks from Northeastern University, and John Triedman from Children's Hospital Boston.

References

[1] M. Alexander, F. Cecchin, E. Walsh et al. J Cardiovasc Electrophysiol, vol. 15, pp. 72-76, 2004.
[2] J. D. Kugler and C. C. Erickson. J Cardiovasc Electrophysiol. 2006 Jan;17(1): 47-48.
[3] G. Ristagno, T. Wang, W. Tang, S. Sun, C. Castillo, and M. H. Weil. Critical Care Medicine, 36(11) (Suppl November): S422–S427, 2008.
[4] M. Jolley, J. Stinstra, D. Weinstein, S. Pieper, S. J. Estepar, F. Cecchin, D. H. Brooks, R. MacLeod, and J. Triedman. HRS Meeting, 2007.
[5] M. Jolley, J. Stinstra, S. Pieper, R. MacLeod, D. H. Brooks, F. Cecchin, and J. Triedman. Heart Rhythm, 5 (No. 4, April 2008):565–572, 2008.
[6] M. Jolley, J. Stinstra, J. Tate, S. Pieper, R. MacLeod, L. Chu, P. Wang, and J. Triedman. Heart Rhythm, 7 (No. 5, May 2010):692–698, 2010.
[7] R. L. Lux, C. R. Smith, R. F. Wyatt, and J. A. Abildskov. IEEE Transactions on Biomedical Engineering, BME-25, No. 3 (May 1978): 270–276, 1978.
[8] JG Stinstra, MA Jolley, JD Tate, DH Brooks, JK Triedman, and RS MacLeod. Comp. in Cardiology. 2008
[9] N. Trayanova, K. Skouibine, and F. Aguel. Chaos, 12(3):962–972, September 2002

www.sci.utah.edu
THE UNIVERSITY OF UTAH